Intratumoral PH-109 INTASYL™ self-delivering RNAi targeting connective tissue growth factor (CTGF) provides efficacy in vivo in a mouse model of metastatic breast cancer

Abstract: 1431

Benjamin Cuiffo, Melissa Maxwell, Dingxue Yan, Andrew Boone, Brianna Rivest, Shenghua Zhou, James Cardia and Simon P. Fricker

SITC 2022
NOVEMBER 8-12 BOSTON MASSACHUSETTS

Abstract

Background Breast cancer is the most diagnosed cancer and the second leading cause of cancer-related deaths globally among women, frequently due to metastatic disease. CTGF orchestrates diverse multicellular processes including embryonic development, wound healing, and tissue repair. CTGF promotes fibrosis in inflammatory diseases and contributes to cancer cell proliferation, migration, invasion, metastasis, and epithelial-mesenchymal transition. High CTGF is associated with poor prognosis in breast cancer. CTGF-inhibition has shown promise in decreasing metastatic dissemination and sensitizing cancer cells to chemotherapy in preclinical models. PH-109 (formerly RXI-109) is a self-delivering RNAi compound built on proprietary INTASYL™ technology, designed to silence human CTGF with high specificity and without need for specialized formulations or drug delivery systems. PH-109 was originally developed and approved as an investigational new drug (IND) for treatment of dermal hypertrophic scarring (Phase 2; NCT02246465) and subretinal fibrosis (Phase 1/2; NCT02599064). Treatment resulted in a statistically significant reduction of CTGF mRNA and protein at the treatment site, with no significant toxicity or adverse effects. Here we present proof-of-concept (POC) in vivo data showing efficacy of intratumorally administered PH-109 in an orthotopic 4T1 model of metastatic mammary cancer.

Methods PH-109 mediated mRNA silencing of CTGF was validated in 4T1 cells in vitro by RT-qPCR. In vivo, 4T1 cells were implanted into the mammary fat pad of BALB/c mice. When tumors reached threshold volume (150 mm³), animals were randomized into treatment groups; test treatments were administered intratumorally (IT) on Days 1, 4, 7, 10 and 13. Vehicle (PBS), a chemically-identical non-targeting control (NTC) INTASYL or PH-109 at two dose concentrations (0.5 mg; 2 mg) were administered IT; doxorubicin chemotherapy (5 mg/kg) was administered intraperitoneally on Days 1, 7, 13. Tumor volumes and body weights were recorded longitudinally. Primary tumors were resected from N = 6/group at ~500 mm³ in survival. Three weeks post-resection, animals were euthanized, and lungs insufflated with India ink and lung macrometastases enumerated. Tumor volumes continued to be recorded for N =6 through study conclusion on Day 19.

INTASYL™ PH-109 Silences Mouse CTGF mRNA in 4T1 Mouse Mammary Cancer Cells in vitro

Figure 1. PH-109 silences murine CTGF mRNA in 4T1 cells in vitro

PH-109 is a self-delivering therapeutic RNAi compound built on Phio's proprietary INTASYL platform, designed to specifically target human CTGF and carrying cross-reactivity towards mouse CTGF. PH-109 mediated on-target concentration dependent silencing of mouse CTGF mRNA compared to PBS vehicle treated control (UTC) or chemically identical but non-targeting INTASYL (NTC) in 4T1 mouse mammary cancer cells. Means ± SEM (n = 3) at 72 h post-treatment are shown. Means were compared to UTC by one way ANOVA and Dunnett's post test. *p<0.05.

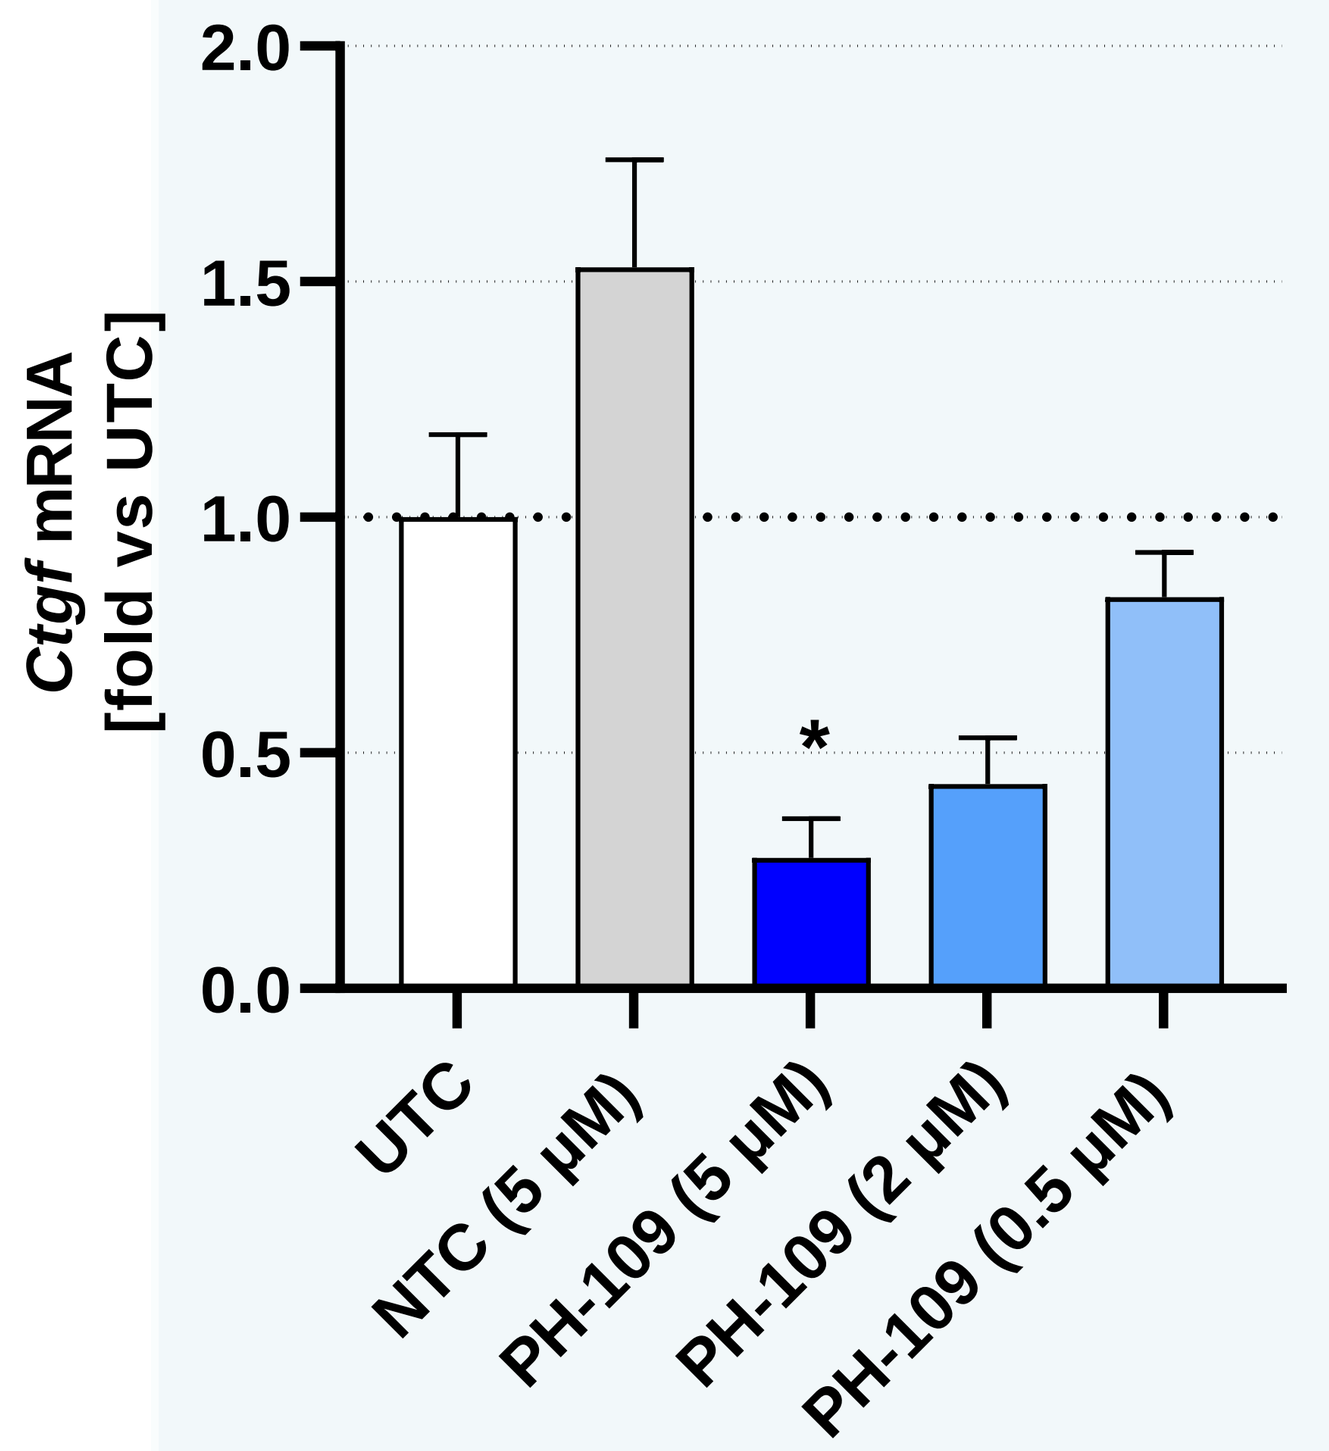
Ctgf mRNA [fold vs UTC]
2.0
1.5
1.0
0.5
0.0
*
UTC
NTC (5 μM)
PH-109 (5 μM)
PH-109 (2 μM)
PH-109 (0.5 μM)

Figure 2. Schematic of in vivo model: local treatment with PH-109 in the in vivo 4T1 model of metastatic mammary cancer

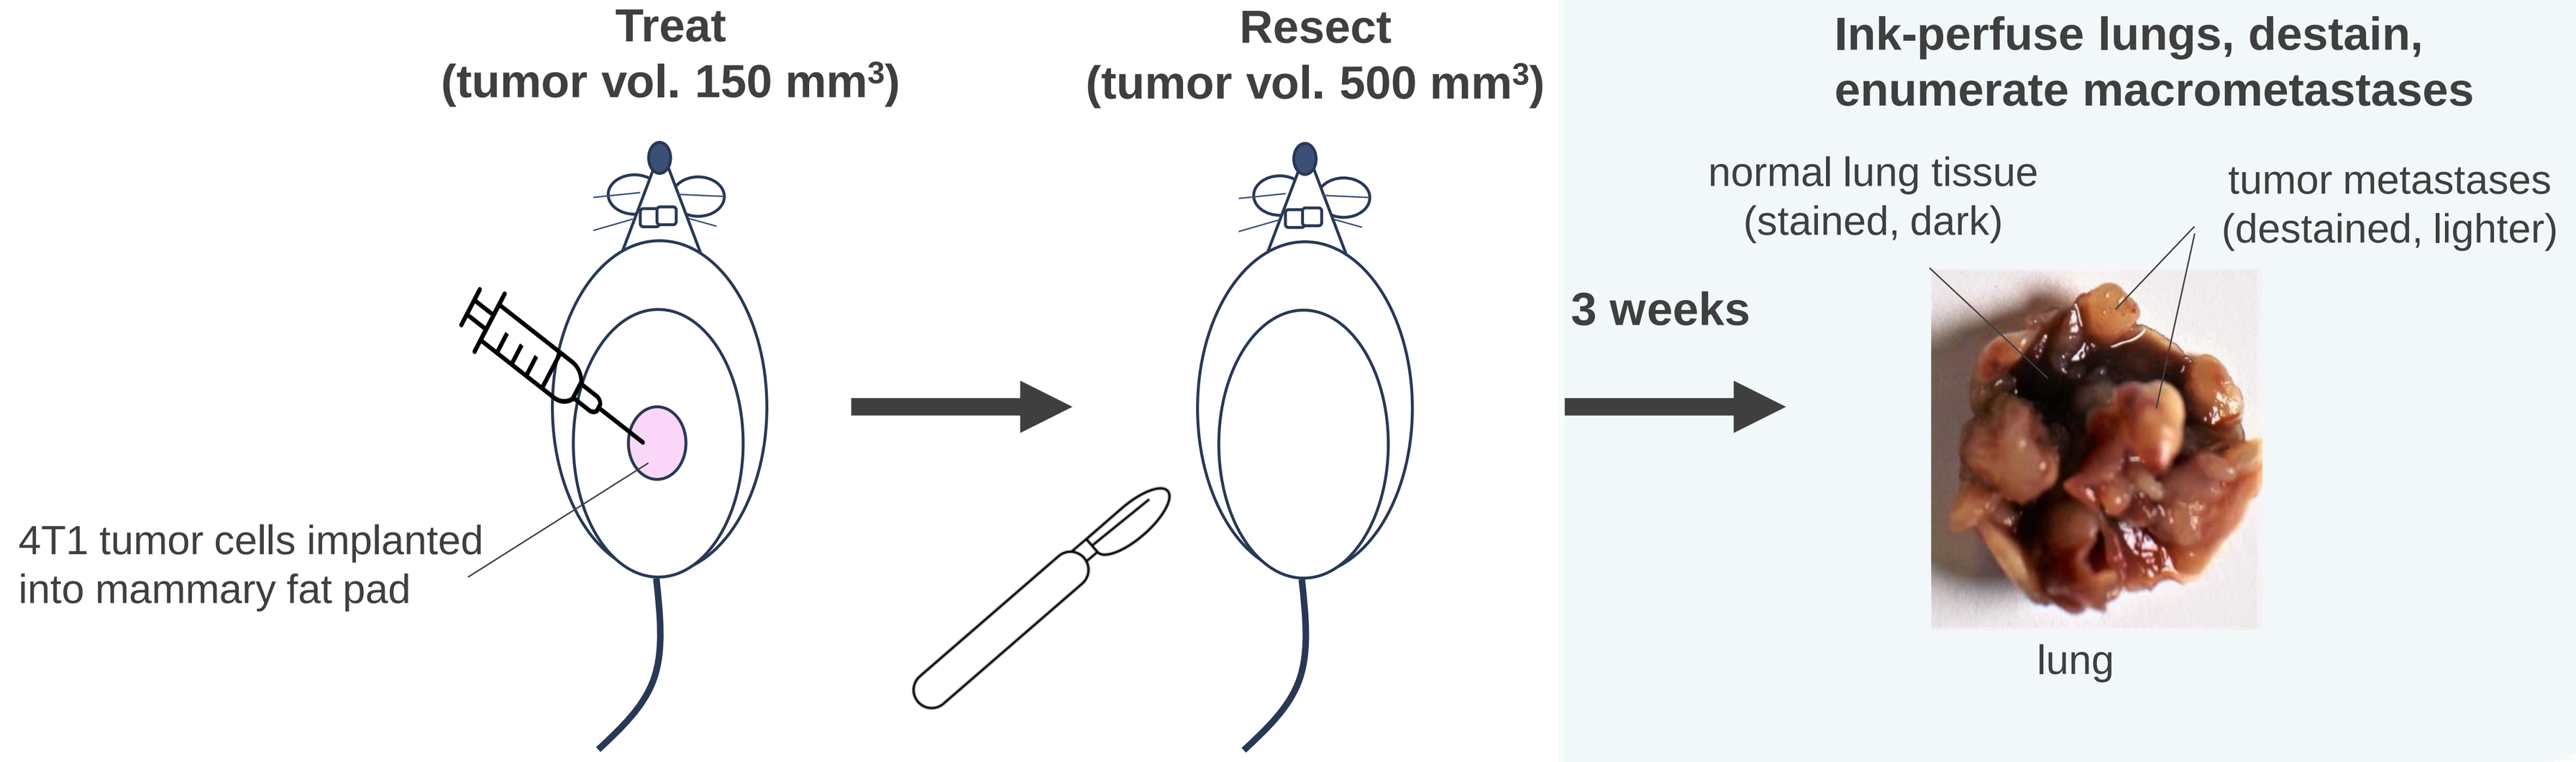
Treat
(tumor vol. 150 mm³)
Resect
(tumor vol. 500 mm³)
Ink-perfuse lungs, destain, enumerate macrometastases
normal lung tissue (stained, dark)
tumor metastases (destained, lighter)
3 weeks
4T1 tumor cells implanted into mammary fat pad
lung

Property of Phio Pharmaceuticals

Intratumoral PH-109 Provides Antitumor Efficacy in an Orthotopic 4T1 Model of Murine Metastatic Mammary Carcinoma

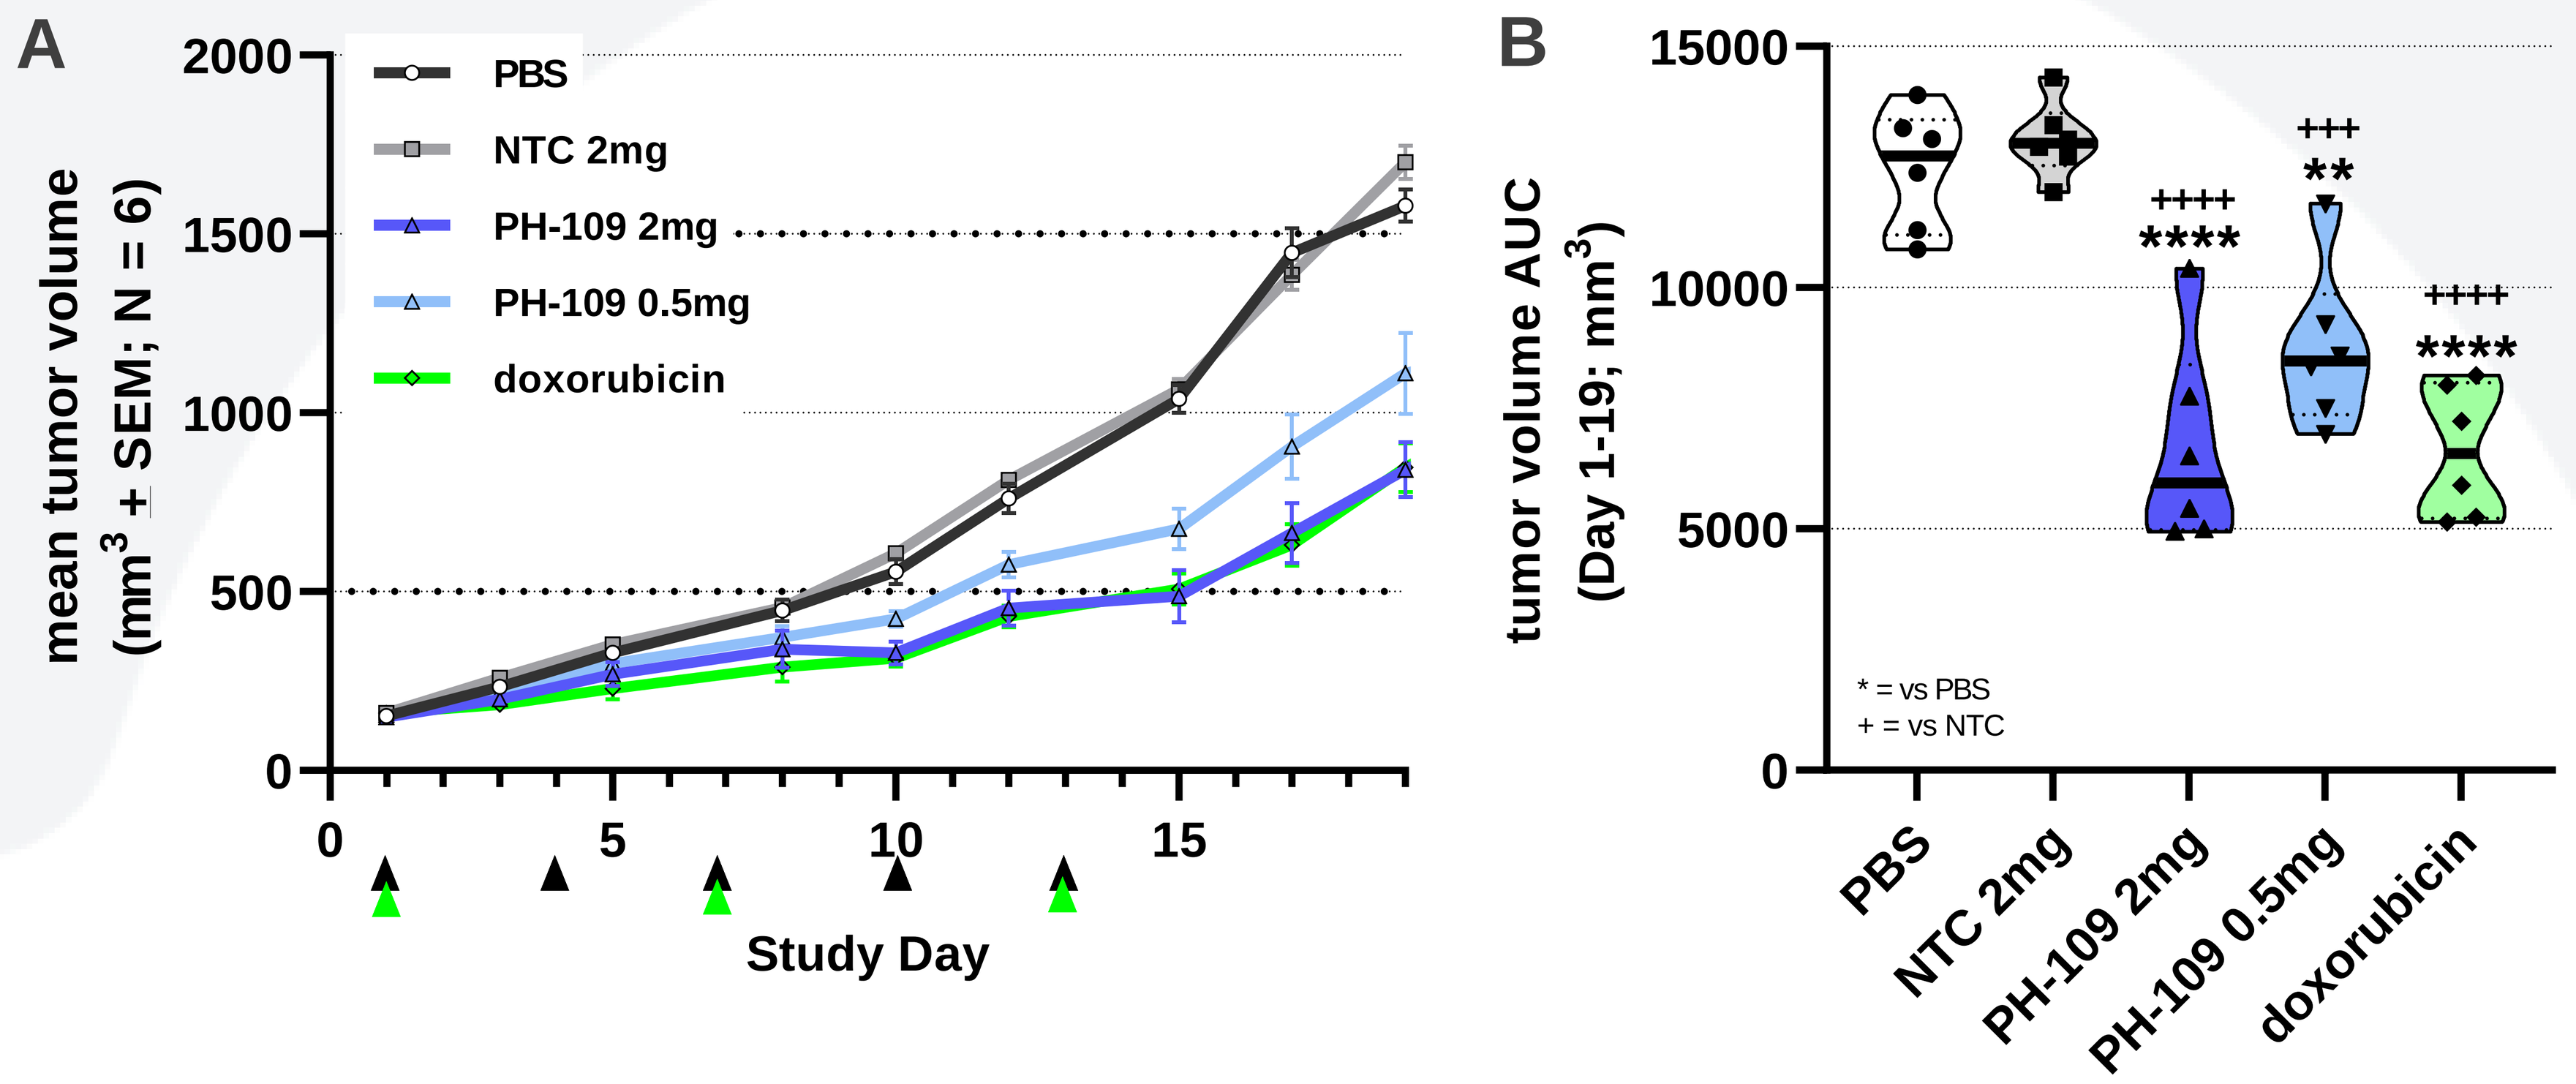
A
mean tumor volume (mm³ + SEM; N = 6)
2000
1500
1000
500
0
PBS
NTC 2mg
PH-109 2mg
PH-109 0.5mg
doxorubicin
0
5
10
15
Study Day
B
tumor volume AUC (Day 1-19; mm³)
15000
10000
5000
0
++++

+++
**
++++

* = vs PBS
+ = vs NTC
PBS
NTC 2mg
PH-109 2mg
PH-109 0.5mg
doxorubicin

Figure 3. Intratumoral (IT) PH-109 provides dose-associated primary tumor growth inhibition in an orthotopic 4T1 model of murine metastatic mammary carcinoma

4T1 tumors were seeded in the mammary fat pad. Tumor volume was assessed 3x/week under treatment (arrows) with IT PH-109 (0.5 or 2 mg / dose; q3d through Day 13) compared to IT vehicle (PBS) or IT NTC. IP doxorubicin (5 mg/kg; IP; q6d through D13) served as positive control arm. **A.** Mean tumor volume (mm³ ± SEM; N = 6) over time. **B.** Cumulative response was assessed by calculating tumor volume area under the curve (AUC) for each animal (Day 1-19). Violin plots with medians and individual animals indicated. Statistical significance assessed by one way ANOVA and Tukey's multiple comparisons *post-hoc* tests. ****p < 0.0001 ***p < 0.001, **p < 0.01. * = vs PBS. + = vs NTC.

Unlike Doxorubicin Treatment, IT PH-109 Does Not Cause Weight Loss

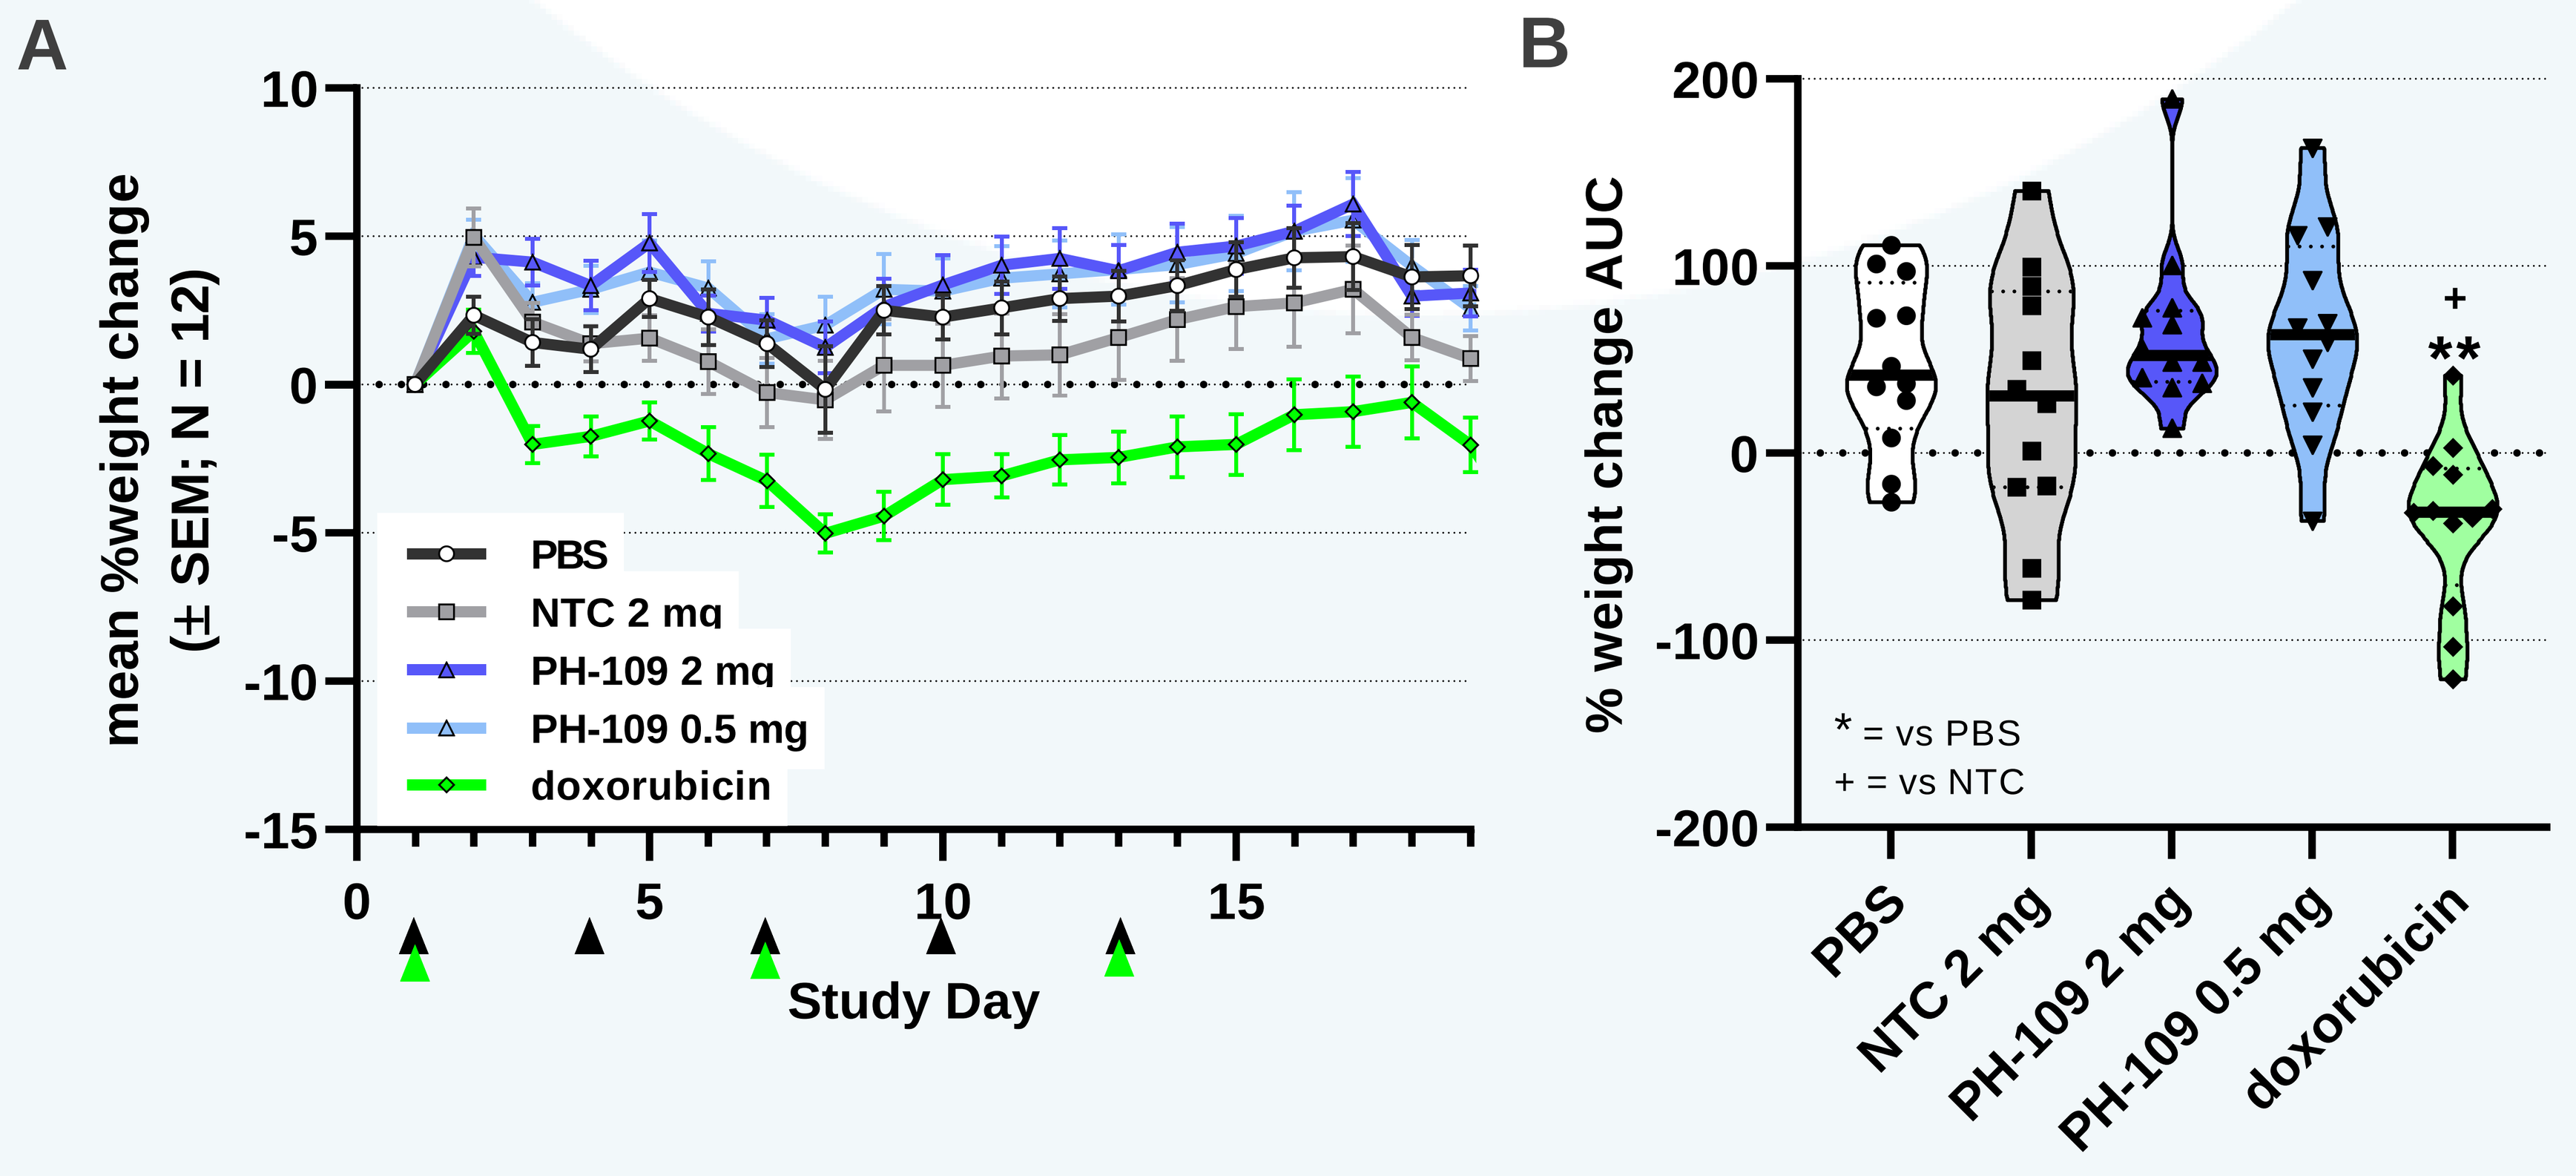
A
mean %weight change (± SEM; N = 12)
10
5
0
-5
-10
-15
PBS
NTC 2 mg
PH-109 2 mg
PH-109 0.5 mg
doxorubicin
0
5
10
15
Study Day
B
% weight change AUC
200
100
0
-100
-200
+
**
* = vs PBS
+ = vs NTC
PBS
NTC 2 mg
PH-109 2 mg
PH-109 0.5 mg
doxorubicin

Figure 4. Unlike IP doxorubicin, IT PH-109 <u>does not</u> elicit weight loss at doses conveying similar efficacy toward primary tumor

Percentage (%) body weight change in the orthotopic 4T1 model. Animals were weighed daily. **A.** Mean % weight change (± SEM; N = 12) over time. **B.** % body weight change AUC (Day 1-19). Violin plots with medians and individual animals are indicated. Statistical significance assessed by one way ANOVA and Tukey's multiple comparisons *post-hoc* tests. **p < 0.001, *p < 0.01. * = vs PBS. + = vs NTC.

in vivo studies were performed at Pharma Models LLC, Marlborough, MA

IT PH-109 Treatment Before Surgery Suppresses up to ~90% of Lung Metastases Following Primary Tumor Resection

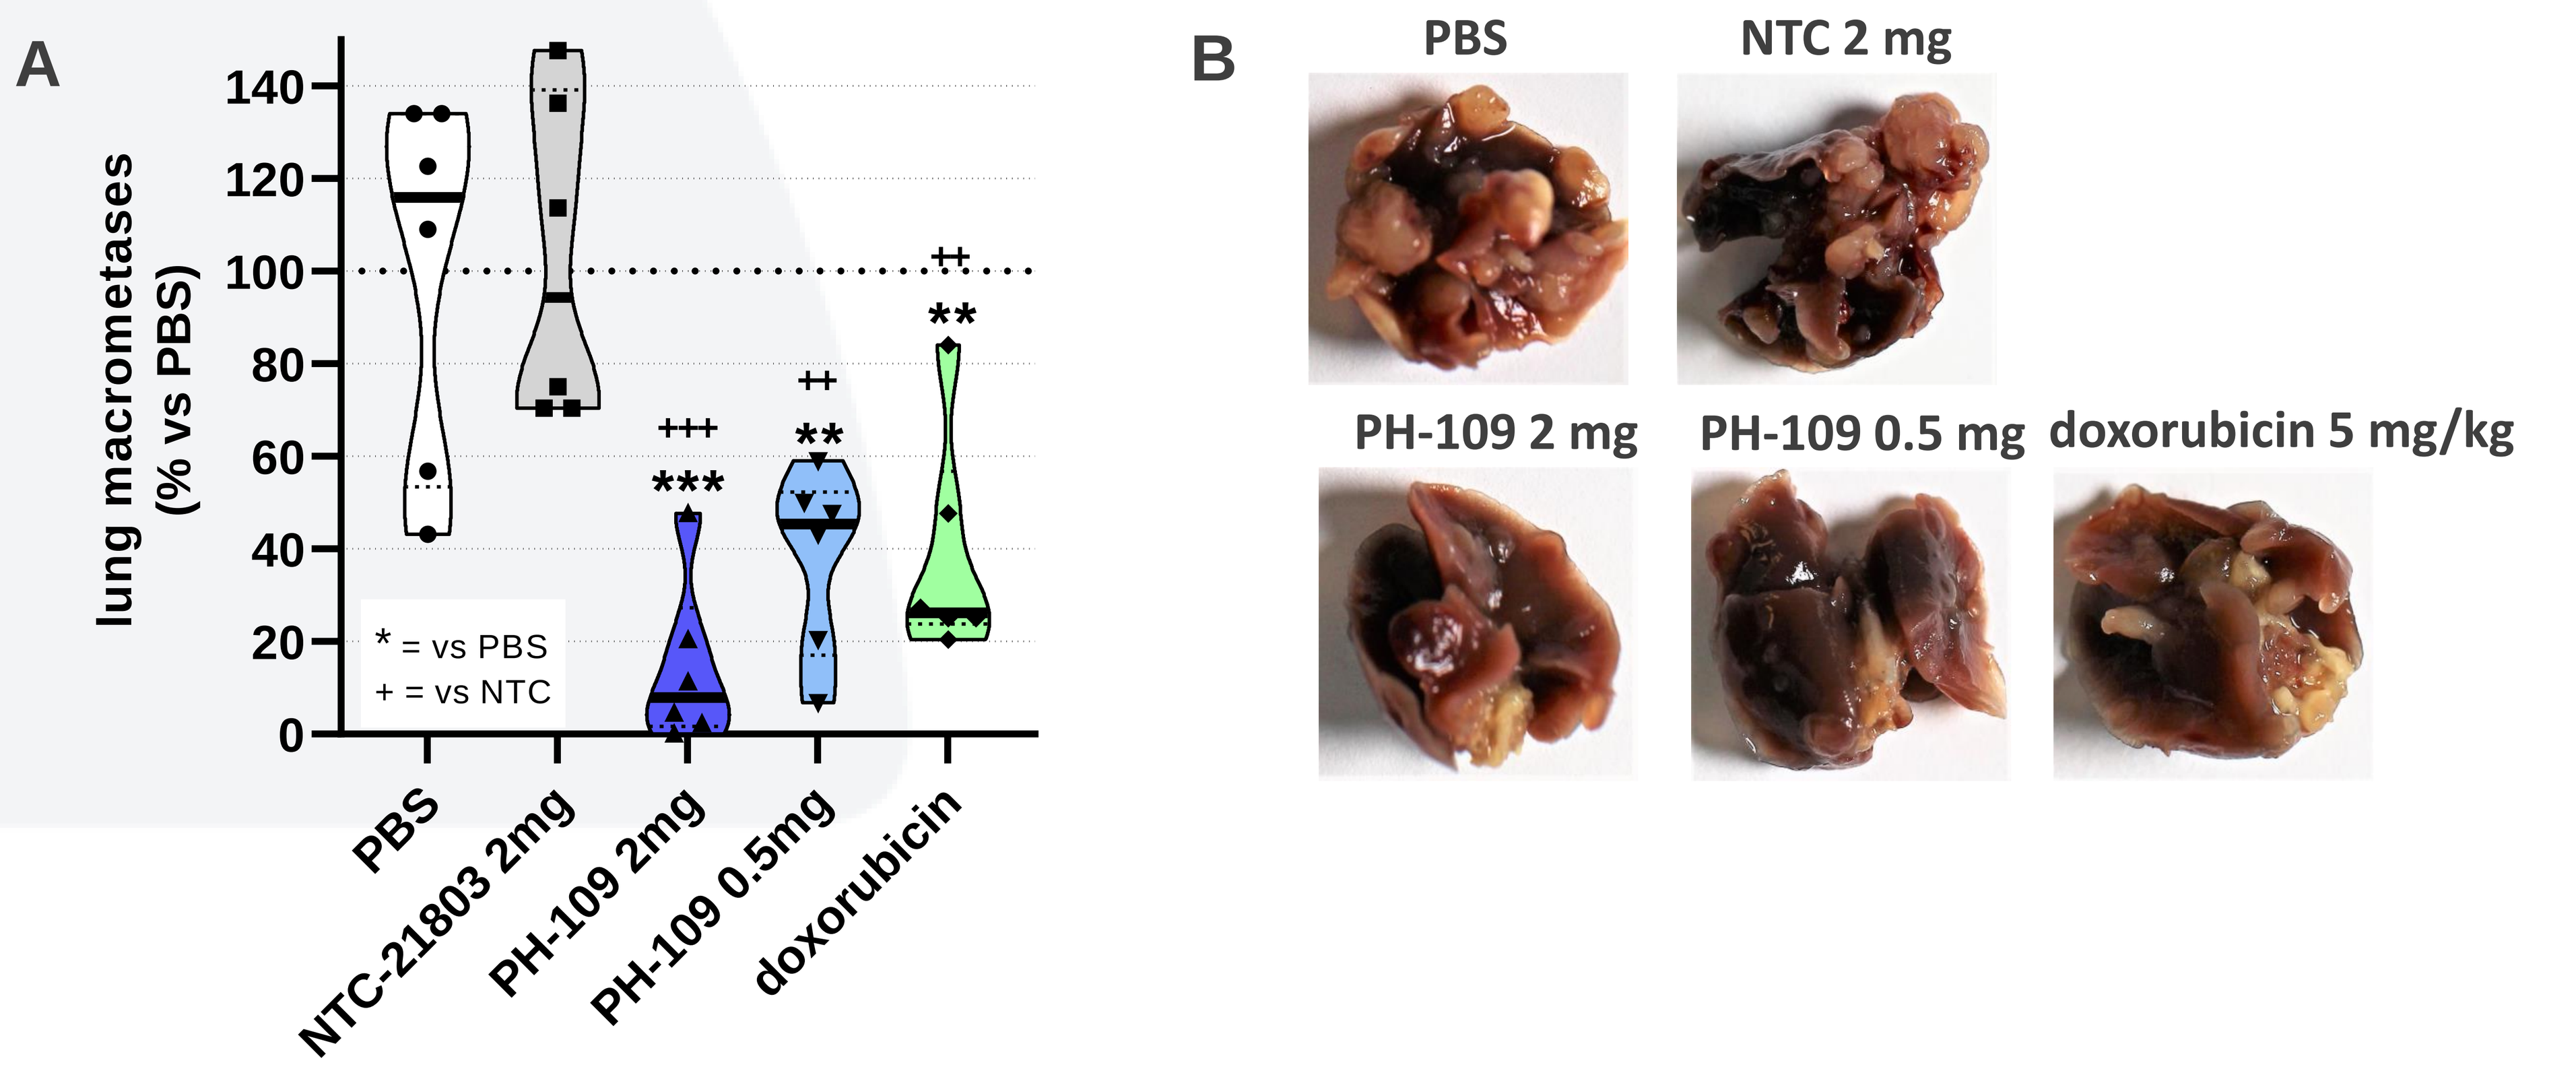
A
lung macrometases (% vs PBS)
140
120
100
80
60
40
20
0
+++

++
**
++
**
* = vs PBS
+ = vs NTC
PBS
NTC-21803 2mg
PH-109 2mg
PH-109 0.5mg
doxorubicin
B
PBS
NTC 2 mg
PH-109 2 mg
PH-109 0.5 mg
doxorubicin 5 mg/kg

Figure 5. IT PH-109 suppresses lung metastases in the 4T1 orthotopic model of metastatic mammary carcinoma

A cohort (n = 6) of animals bearing mammary fat pad 4T1 tumors had their tumors resected as each tumor reached a threshold volume of 500 mm³. Three weeks after each resection, lungs were isolated and lung macrometastases enumerated (both faces of each lung). **A.** Percentage (%) of lung macrometastases for each treatment group compared to treatment with PBS. Violin plots with medians and individual animals are indicated Statistical significance assessed by one way ANOVA and Tukey's multiple comparisons *post-hoc* tests. ***p < 0.001, **p < 0.01. * = vs PBS; + = vs NTC. **B.** Representative lung photos by treatment group. Tumor macrometastases nodules appear lighter compared to normal lung tissue.

Summary and Conclusions

- **PH-109 conveyed on-target concentration dependent silencing of mouse CTGF mRNA in 4T1 cells in vitro.**
- **Intratumoral (IT) PH-109 provided dose-associated primary tumor growth inhibition in an orthotopic 4T1 model of murine metastatic mammary carcinoma**
- **IT PH-109 conveyed similar efficacy toward primary tumor but without the toxicity (denoted by weight loss) associated with systemic doxorubicin chemotherapy.**
- **IT PH-109 treatment given prior to surgery robustly suppressed lung metastases.**
- **These studies provide proof-of-concept in vivo antitumor efficacy for IT INTASYL PH-109 targeting CTGF. PH-109 was previously evaluated in over 150 subjects without significant toxicity.**
- **These data in a clinically relevant orthotopic mouse model of metastatic breast cancer could support accelerated clinical investigation of PH-109 as an anticancer therapeutic.**