A Phase 1b study to evaluate the safety, tolerability, pharmacokinetics and anti-tumor activity of neoadjuvant use of PH-762 administered intratumorally in subjects with advanced melanoma

Caroline Robert[1], François-Xavier Danloss[1], Emilie Routier[1], Benjamin Cuiffo[2], James Cardia[2], Simon P. Fricker[2]

[1]Oncology Department, Gustave Roussy, and Paris-Saclay University, Villejuif, France
[2]Phio Pharmaceuticals, Marlborough, MA, United States

Abstract:788

SITC 2022 NOVEMBER 8-12 BOSTON MASSACHUSETTS

Abstract

Background Immunotherapy with antibodies targeting PD-1 and CTLA-4 has shown significant benefit in late-stage melanoma. However, as response is limited to approximately 60%, and with emergence of resistance, further improvements in therapeutic options are still required. Two approaches for improving the outcome of immunotherapy with checkpoint inhibitors are neoadjuvant treatment and local intratumoral (IT) injection. Neoadjuvant immunotherapy can induce significant pathological responses that seem to be associated with a decrease in the risk of relapse. Currently there is no neoadjuvant standard of care for patients with resectable, advanced melanoma. IT immunotherapy uses the tumor as its own vaccine to activate the immune system, priming an anti-tumor immune response and minimizing systemic exposure and off-target toxicities. PH-762 is a potent RNAi molecule targeting PD-1 that contains structural and chemical modifications conferring rapid and efficient tissue uptake suitable for IT administration. In pharmacology studies PH-762 provided robust silencing of PD-1 associated with T cell activation, and dose-dependent inhibition of tumor growth in *in vivo* syngeneic tumor models with on-target PD-1 silencing-associated with immunostimulatory effects in the tumor microenvironment.

Methods The primary objective of this first-in-human study is to evaluate the safety of neoadjuvant use of PH-762 administered by IT injection. Secondary objectives include PK after IT injection, potential immunologic and pathologic tumor responses, and determination of the recommended Phase 2 dose. Subjects must have histologically confirmed stage IIIB/IIIC/IIID or IV oligometastatic cutaneous melanoma with at least one resectable lesion that is large enough to allow IT injection, and that can undergo repeated biopsy. Subjects with brain metastases, leptomeningeal disease, uveal melanoma, and auto-immune disease are excluded. PH-762 is administered IT into one designated tumor lesion once weekly for 4 weeks prior to surgical excision at 5-6 weeks after the initial injection. The dose of PH-762 is normalized to tumor volume to ensure an equivalent local dose (tumor tissue concentration). Post tumor excision, subjects are followed-up for 6 weeks. Primary endpoint is determination of a safe dose of PH-762 assessed by incidence of dose limiting toxicities (DLT) prior to tumor resection. Bayesian optimal interval (BOIN) design will be employed to evaluate escalating doses of PH-762 with up to 5 dose levels in cohorts of 3 or more subjects. Tumor changes will be evaluated per RECIST criteria (version 1.1 and iRECIST adapted for IT therapy) and pathological response. Secondary endpoints include immunological response in tumor tissue and blood. The first cohort is currently being enrolled.

PHIO-762-2101- Study Design: A First-in-Human Phase 1b study

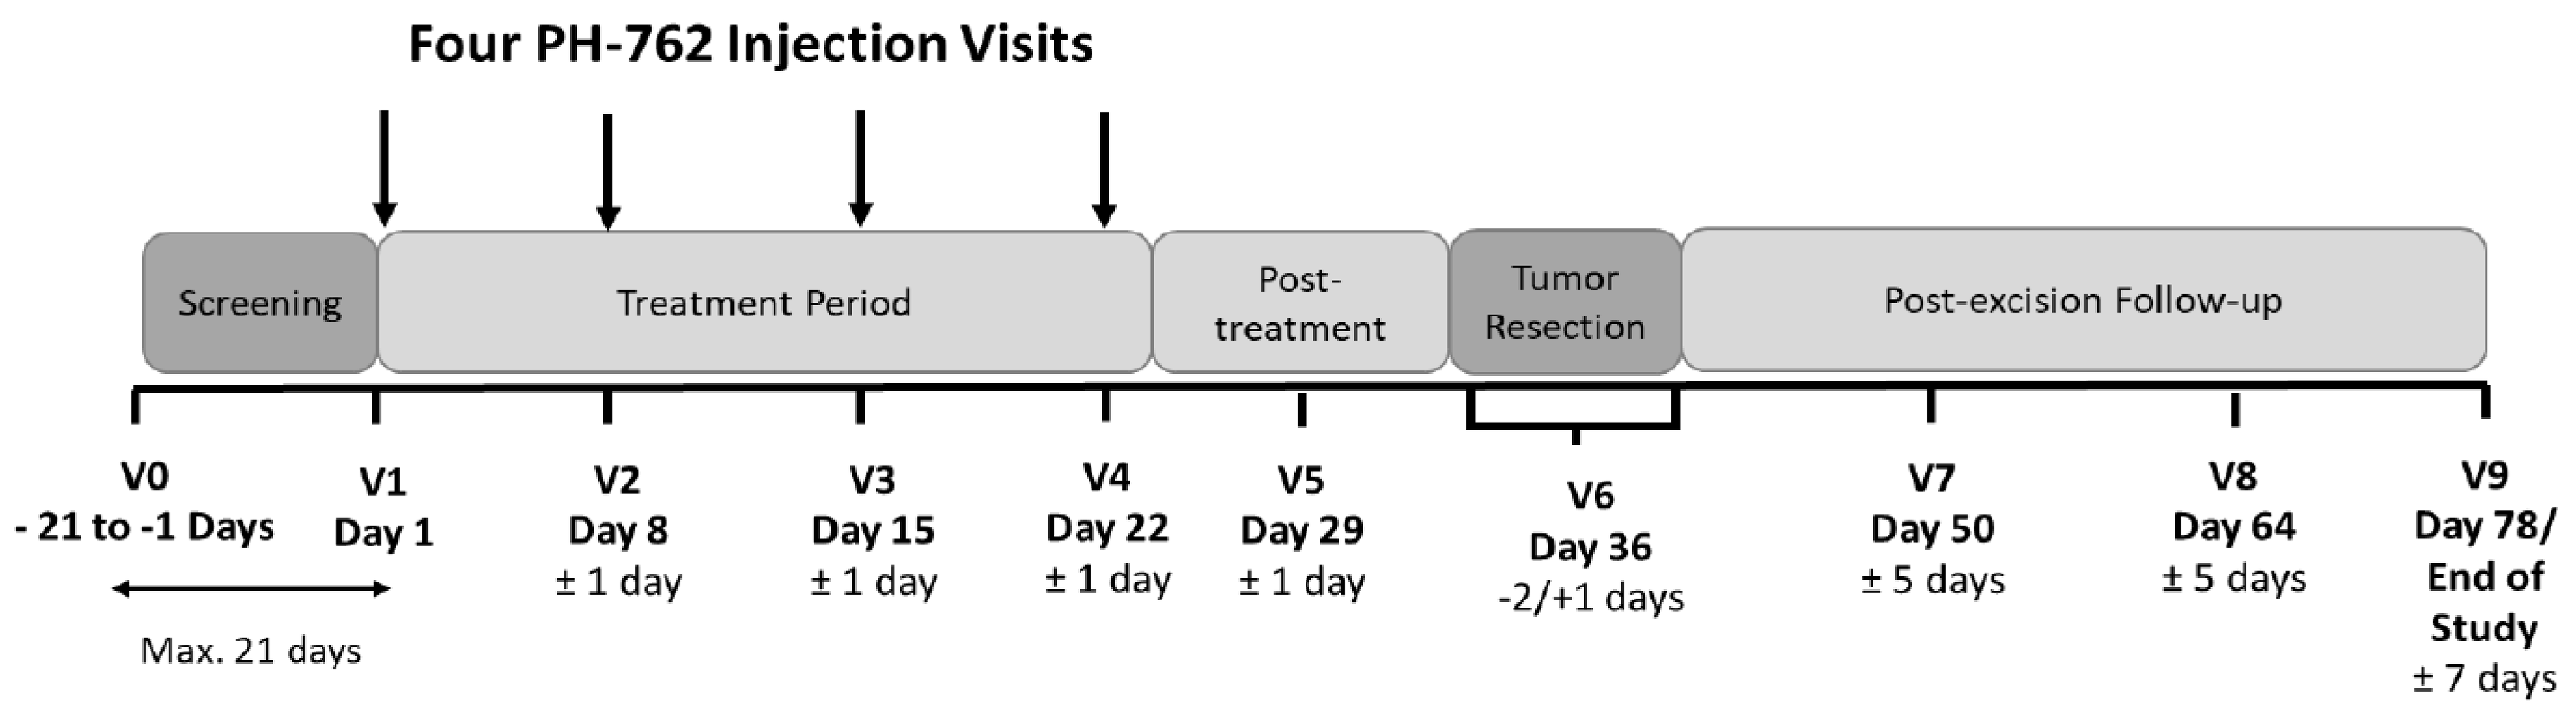
Four PH-762 Injection Visits
Screening
Treatment Period
Post-treatment
Tumor Resection
Post-excision Follow-up
V0
- 21 to -1 Days
Max. 21 days
V1
Day 1
V2
Day 8
± 1 day
V3
Day 15
± 1 day
V4
Day 22
± 1 day
V5
Day 29
± 1 day
V6
Day 36
-2/+1 days
V7
Day 50
± 5 days
V8
Day 64
± 5 days
V9
Day 78/
End of
Study
± 7 days

Primary Objective: Determine a safe dose of PH-762 administered by IT injection in subjects with stage IIIB/IIIC/IIID or IV resectable melanoma

Overview:

- Single center, up to 21 subjects
- Subjects in all cohorts will receive standard adjuvant treatment as per investigator choice
- Up to 5 dose levels will be tested (dose administration based on dose limiting toxicity (DLT))

Property of Phio Pharmaceuticals

Study Synopsis

Primary endpoint

Incidence of DLT prior to tumor resection.

Secondary endpoints

Pharmacokinetics (PK) of PH-762 after IT injection

Tumor response evaluated per RECIST criteria (version 1.1 and pathological response)

Determine a recommended dose of PH-762 in follow-up clinical studies

Immunological response to PH-762 therapy measured by correlative biomarkers in peripheral blood and tumor tissue

Inclusion criteria

Subject ≥ 18 years of age

Histologically confirmed stage IIIB/IIIC/IIID or IV oligometastatic cutaneous melanoma (with at least one injectable and resectable lesion)

At least one resectable melanoma deposit that is large enough to allow IT injections (≥ 1 cm in diameter and ≥ 1.5 cm for lymph nodes), but smaller than 3 cm

Medically fit enough to undergo surgery as determined by the treating medical and surgical oncology teams

Exclusion criteria

Subjects with brain metastases or leptomeningeal disease, or primary tumors of uveal melanoma are excluded

Any serious or uncontrolled medical disorder including auto-immune disease

Prior malignancy active within the previous 3 years

Receiving cancer therapy

Bayesian Optimal Interval (BOIN) Design for dose escalation/ de-escalation: BOINN Flow Chart

The BOIN design is implemented in a simple way similar to the traditional 3+3 design but provides a more efficient statistical estimate of the MTD

The BOIN design uses the following rule:

- If the observed DLT rate at the current dose is ≤ 0.236 escalate the dose to the next higher dose level
- If the observed DLT rate at the current dose is > 0.359, de-escalate the dose to the next lower dose level
- Within 0.239 – 0.358 stay at the current dose.

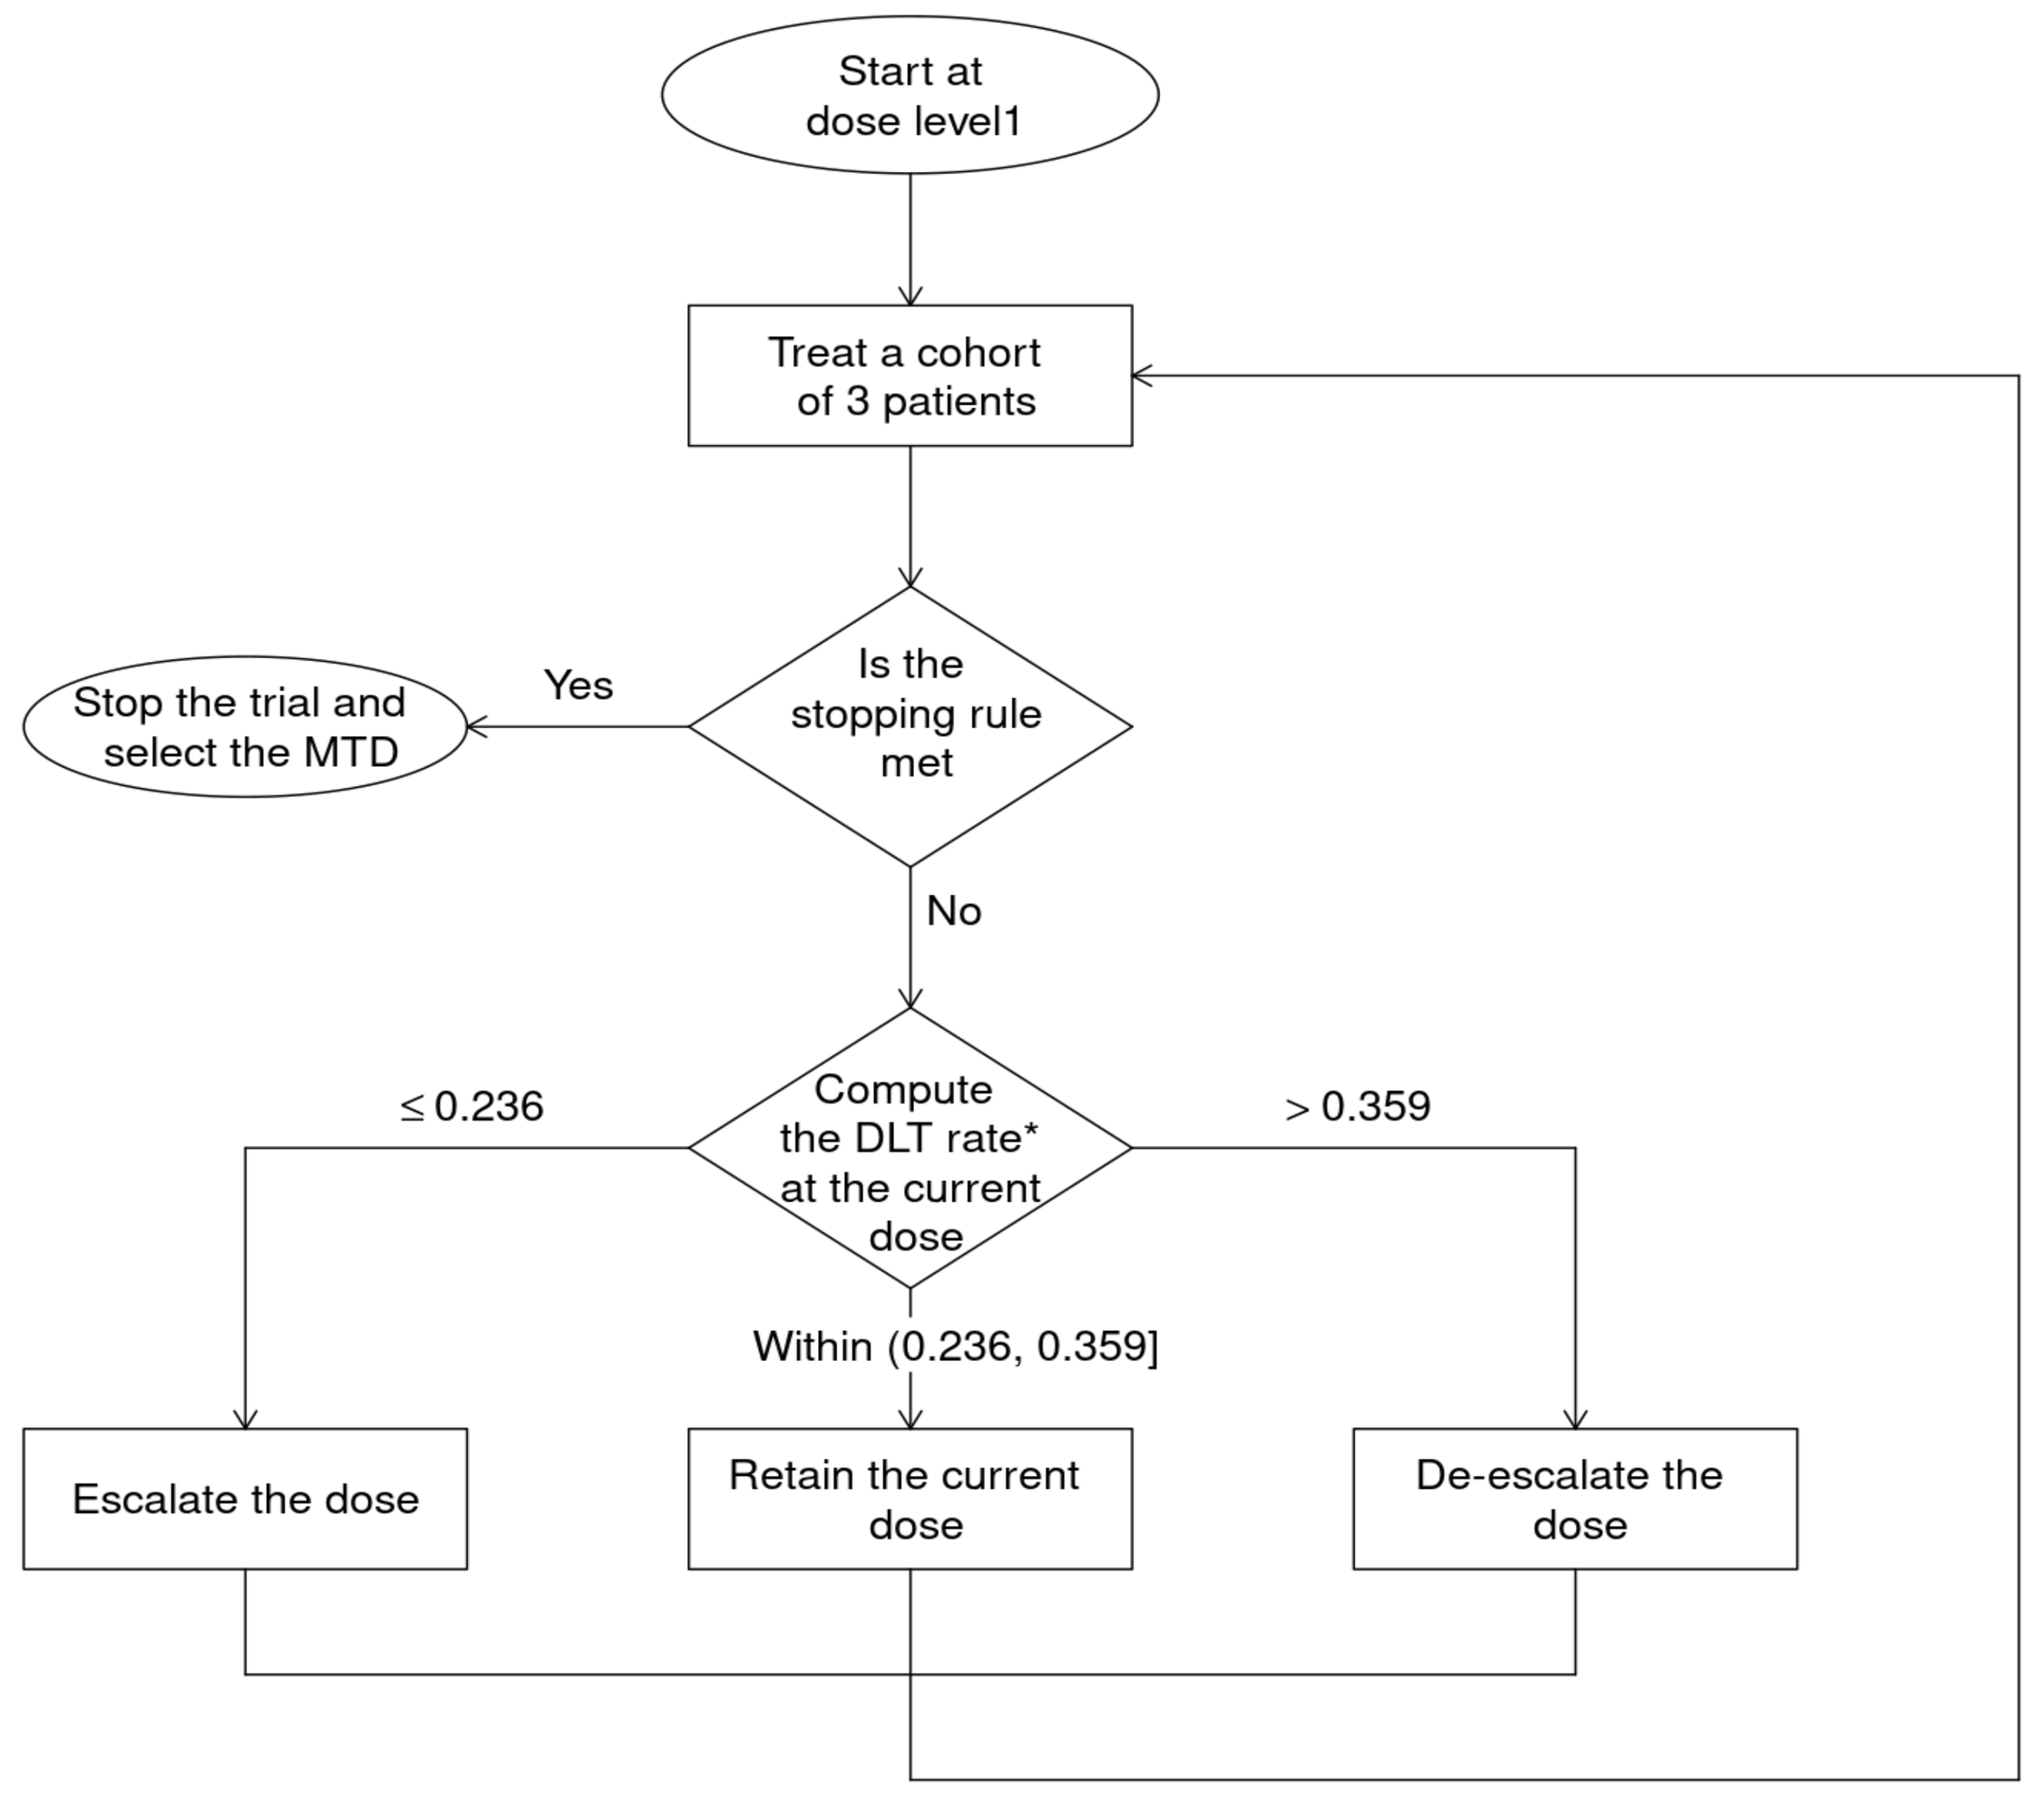
Start at dose level1
Treat a cohort of 3 patients
Is the stopping rule met
Yes
Stop the trial and select the MTD
No
Compute the DLT rate* at the current dose
≤ 0.236
> 0.359
Within (0.236, 0.359]
Escalate the dose
Retain the current dose
De-escalate the dose

$$\text{* DLT rate} = \frac{\text{Total number of patients who experienced DLT at the current dose}}{\text{Total number of evaluable patients treated at the current dose}}$$

Overview of Dosing Based on Dose Level Cohort Concentration and Tumor Size

		Tumor size: 1 cm	1.5 cm	2 cm	2.5 cm	3 cm	Targeted Tumor tissue concentration
Dose level 1	1.14 mg/ml	0.29 mg	1.14 mg	2.28 mg	4.56 mg	7.70 mg	0.57 mg / cm³
Dose level 2	2.4 mg/ml	0.60 mg	2.39 mg	4.78 mg	9.56 mg	16.13 mg	1.19 mg / cm³
Dose level 3	5.0 mg/ml	1.25 mg	5.01 mg	10.02 mg	20.03 mg	33.81 mg	2.50 mg / cm³
Dose level 4	10.5 mg/ml	2.62 mg	10.50 mg	20.99 mg	41.98 mg	70.86 mg	5.25 mg / cm³
Dose level 5	22.0 mg/ml	5.50 mg	22.00 mg	44.00 mg	88.00 mg	148.52 mg	11.0 mg / cm³

The administered dose of PH-762 will be normalized to tumor volume to ensure equivalent local dose administration (tumor tissue concentration)

The five dose concentrations are:

- Dose level 1: 1.14 mg/mL to obtain a tumor tissue concentration of 0.57 mg/cm³
- Dose level 2: 2.39 mg/mL to obtain a tumor tissue concentration of 1.19 mg/cm³
- Dose level 3: 5.01 mg/mL to obtain a tumor tissue concentration of 2.50 mg/cm³
- Dose level 4: 10.50 mg/mL to obtain a tumor tissue concentration of 5.25 mg/cm³
- Dose level 5: 22.00 mg/mL to obtain a tumor tissue concentration of 11.00 mg/cm³

Perspective and Key Messages

- **Neoadjuvant immunotherapy can induce significant pathological responses that seem to be associated with a decrease in the risk of relapse**
- **Currently there is no neoadjuvant standard of care for patients with resectable, advanced melanoma**
- **Neoadjuvant treatment with PH-762 RNAi provides an alternative therapeutic modality for patients with resectable, advanced melanoma**
- **Direct administration of PH-762 to the tumor, in mouse models, can prime a local immune response by silencing PD-1 in the tumor microenvironment and generate a systemic immune response**
- **IT administration has the potential to minimize systemic exposure and off-target toxicity**
- **Enrollment is ongoing for the first cohort of PHIO-762-2101**

Approval was provided by: Comite de Protection de Personnes Sud Mediterranee III, 2021.12.04 bis _21.04174.000055.

EudraCT number 2021-002859-10.